PHASE 1 STUDIES OF THE SAFETY, PHARMACOKINETICS AND PHARMACODYNAMICS OF AT2101, A PHARMACOLOGICAL CHAPERONE FOR THE TREATMENT OF GAUCHER DISEASE

David J. Palling[1], Karin Ludwig[1], Mara Casar[1], Sheela Sitaraman[1], Chris Duke[1], Alan S. Marion[2], Mark Stiles[2], Stacey Duey[2], Tommy Tsikos[3], David Goblot[3], Malle Jurima-Romet[3], Michel Roy[3], Marika DiMarco[3], and Rahma Demnati[3].

[1]Amicus Therapeutics, 6 Cedarbrook Drive, Cranbury, NJ, 08512; [2]MDS Pharma Services, 621 Rose St., Lincoln, NE 68502; [3]MDS Pharma Services, 2350 Rue Cohen, Montreal, Quebec H4R 2N6

Abstract

AT2101 (isofagomine tartrate) is a pharmacological chaperone in development for the treatment of the lysosomal storage disorder Gaucher disease. Using cell-based and animal models we have shown that AT2101 increases cellular levels of β-glucocerebrosidase (GCase), the enzyme deficient in Gaucher disease. Randomized double-blind phase 1 clinical studies were performed in 72 healthy volunteers, (38 male, 34 female). AT2101 was orally administered as an aqueous solution.

In a first-in-human single ascending dose study, doses of 8, 25, 75, 150 (two cohorts), and 300 mg were administered (6 active, 2 placebo in each cohort). In a multiple ascending dose study, doses of 25, 75, and 225 mg were administered daily for 7 days (6 active, 2 placebo in each cohort). In both studies, AT2101 was generally well tolerated at all doses and treatment-emergent adverse events in both studies were mostly mild. No serious adverse events occurred.

AT2101 showed good systemic exposure via the oral route. In the single-dose study, plasma AUC and Cmax values were linearly correlated with administered dose. Mean plasma levels peaked at 3.4 hours. (SEM: 0.6 hours.) and the plasma elimination half-life was 14 hours. (SEM: 2 hours.). In the multiple-dose study, after 7 days of oral administration, the pharmacokinetic behavior was found to be linear with dose, with no unexpected accumulation of AT2101. Tmax and half-life values were similar to those observed in the single-dose study.

In the multiple-dose study, GCase activity in isolated white blood cells was measured at days 1, 3, 5 and 7 during administration of AT2101, and at days 9, 14 and 21 during the post-treatment washout period. In all subjects receiving AT2101 there was a marked increase in GCase levels during the AT2101 treatment period, followed by a decrease upon removal of the drug and a return to near baseline levels by day 21. The increase in enzyme level was dose-related, reaching approximately 3.5-fold above baseline levels. These results for the safety, pharmacokinetics and preliminary pharmacodynamic effects in healthy volunteers support the further evaluation of AT2101 for the treatment of Gaucher disease.

Introduction

Gaucher disease is an autosomal recessive lysosomal storage disorder caused by a deficiency in acid β-glucocerebrosidase (GCase) activity. While enzyme replacement and substrate reduction therapies are currently available to treat Gaucher disease, the use of orally administered small molecule pharmacological chaperones to stabilize and increase the activity of mutant proteins is a promising new approach.

AT2101 (isofagomine tartrate) is a pharmacological chaperone in development for the treatment of Gaucher disease. Cell-based and animal model experiments have shown that AT2101 increases cellular levels of GCase and can affect biochemical and clinical markers of disease (ACMG 2007 poster 368). Pre-clinical IND-enabling studies have shown that AT2101 has an acceptable safety profile to allow its use in human clinical trials. In this first phase of clinical evaluation, we have administered the compound to healthy volunteers in two studies under single ascending dose (phase 1a study) and multiple ascending dose (phase 1b study) protocols.

Objectives

The primary objective of both the phase 1a and 1b studies was to evaluate the safety and tolerability of AT2101. Secondary objectives were to gain information on the pharmacokinetics of AT2101, and in the phase 1b study, to evaluate a potential pharmacodynamic response to AT2101 – GCase activity in white blood cell lysate.

Study Design

Both studies were single-center, randomized, double-blind, and placebo-controlled, with AT2101 administered orally under fasting conditions as an aqueous solution to healthy volunteers aged between 19 and 55. In the phase 1a study, a total of 48 subjects were enrolled to evaluate 5 single dose levels: 8, 25, 75, 150 (2 cohorts), and 300 mg. In the phase 1b study, a total of 24 subjects were enrolled to evaluate 3 dose levels administered once per day for 7 days: 25, 75, and 225 mg. In both studies, of the eight subjects in each group; six were randomized to receive AT2101, and two subjects received placebo. Subjects were confined from the evening of day -1 until 24 hours after completion of dosing. In the phase 1a study subjects returned at 48 hours (PK sampling) and 7 days (safety follow-up) following dosing. In the phase 1b study subjects returned at 48 hours (PD sampling), 7 days (PD sampling and safety follow-up), and 14 days (PD sampling) following the last dose.

Demographics

Phase 1a: 48 healthy subjects (24 male and 24 female). Mean age of 27 years (range 19-54 years), mean height of 172 cm (range 157-188 cm), and mean weight of 71.4 kg (range 54.0-93.0 kg).

Phase 1b: 24 healthy subjects (14 male and 10 female). Mean age of 36 years (range 19-55 years), mean height of 175 cm (range 152-191 cm), and mean weight of 76.1 kg (range 52.2-99.3 kg).

Methods

Safety and tolerability objectives were accomplished through evaluation of a number of safety parameters, including clinical signs and symptoms, vital signs, physical examination, ophthalmologic exam, ECG, clinical laboratory tests (hematology, clinical chemistry, urinalysis) and adverse events, which were classified according to MedDRA Version 9.0.

AT2101 in plasma and urine was analyzed using a validated LC-MS/MS method with an analytical range of 2.00 – 400 ng/mL for plasma, and 0.5 – 100 µg/mL for urine.

GCase activity in white blood cell lysates was measured using a qualified fluorescence enzyme assay incorporating 4-methyl umbeliferryl-β-glucoside as the enzyme substrate.

Pharmacokinetic parameters were calculated using standard procedures. In the phase 1a study a full AT2101 PK profile was determined for 48 hours following dosing. In the phase 1b study a full AT2101 PK profile was determined for 24 hours following the first dose, C_{min} values were obtained pre-dose on day 6 and 7, and another full profile was determined for 24 hours following the final dose on day 7. Statistical PK/PD analyses included the power model, the analysis of variance (ANOVA) and Helmert contrasts.

GCase activity was determined pre-dose on days 1, 3, 5 and 7 and at the corresponding time on days 9, 14 and 21.

Safety and Tolerability

Summary

- **AT2101 was well tolerated and there were no withdrawals due to adverse events.**
- **There was a total of 91 adverse events; 74 were considered to be treatment-related.**
- **All AEs were transient and mild or moderate in severity.**

Table 1. Overall Adverse Event Distribution

	Phase 1a Single Dose						Phase 1b Multiple Dose (7-Day)				Totals
Dose (mg)	8	25	75	150	300	Pbo	25	75	225	Pbo	
N	6	6	6	12	6	12	6	6	6	6	72
Total Number of Adverse Events	0	1	3	9	2	5	0	20	36	17	91
Number (%) of Subjects with Adverse Events	0	1 (17%)	3 (50%)	5 (42%)	1 (17%)	2 (17%)	0	6 (100%)	6 (100%)	6 (100%)	30 (42%)

Table 2. Severity and Relatedness of Adverse Events Considered Treatment-Related

	Severity		Treatment Relatedness		
	Mild	Moderate	Probable	Possible	Remote
Phase 1a	15	2	5	10	2
Phase 1b	47	10	0	47	10

Table 3. Incidence of Common (> 5%) Adverse Events Considered Treatment-Related

	Phase 1a Single Dose						Phase 1b Multiple Dose (7-Day)				Totals (% total AEs)
Dose (mg)	8	25	75	150	300	Pla	25	75	225	Pla	
N	6	6	6	12	6	12	6	6	6	6	72
Headache			1	1				6(3)	5(2)	5(4)	18 (20%)
Nausea				2 (2)				3 (2)	1	1	7 (8%)
Dizziness				2 (1)		1		1	1		5 (5%)
Blood Bilirubin Increased			1	3 (3)		1					5 (5%)

Parenthetic values refer to number of subjects reporting AE

Laboratory Abnormalities

The only clinically significant laboratory abnormality was elevated bilirubin, occurring in 5 subjects in the phase 1a study. Thoursee subjects received 150 mg of AT2101, one subject received 75 mg, and one subject received placebo. Elevation was observed 24 hours post dose in the four subjects receiving AT2101 and 12 hours post dose in the subject receiving placebo. The AE was transitory; bilirubin values had returned to normal upon recheck after 12 hours in the placebo subject and 5 days in the AT2101-treated subjects (no earlier re-checks in these subjects). The elevations were considered mild and probably treatment- related. The elevated values ranged from 1.7 to 2.4 mg/dL (upper limit of normal: 1.6 mg/dL).

Pharmacokinetics: Single Ascending Dose

Table 4. Pharmacokinetic Results for AT2101 in Plasma and Urine following a single oral dose

PK Parameter	Dose (mg)					
	8	25	75	150	150	300
AUC_{inf} (ng·h/mL)	102 (45)	362 (32)	1199 (26)	2224 (44)	1643 (44)	3464 (23)
C_{max} (ng/mL)	11.3 (59.4)	42.9 (38.4)	130.3 (36.5)	235.0 (38.6)	150.9 (28.7)	384.0 (25.7)
t_{max} (h)	3.50 (3.00 – 4.00)	3.00 (1.50 – 4.00)	3.50 (2.00 – 4.00)	3.50 (3.00 – 6.00)	3.50 (1.50 – 5.00)	3.09 (1.50 – 5.00)
$t_{½}$ (h)	5.14 (1.44)	9.00 (1.33)	16.1 (10.6)	19.9 (11.7)	18.3 (4.3)	19.9 (9.1)
CL/F (L/h)	42.0 (17.9)	35.7 (11.2)	31.8 (7.8)	35.7 (13.6)	48.6 (20.2)	43.8 (9.2)
V_{area}/F (L)	308 (134)	465 (171)	715 (458)	1181 (1171)	1281 (719)	1189 (376)
F_e (%)	7.0 (7.6)	13.4 (10.7)	13.4 (5.68)	14.2 (6.67)	10.1 (4.43)	11.6 (3.31)

AUC and C_{max} results are geometric mean (CV%), t_{max} results are median (min-max). All other results are arithmetic mean ± SD

Figure 1. Mean plasma concentrations of AT2101 following a single oral dose

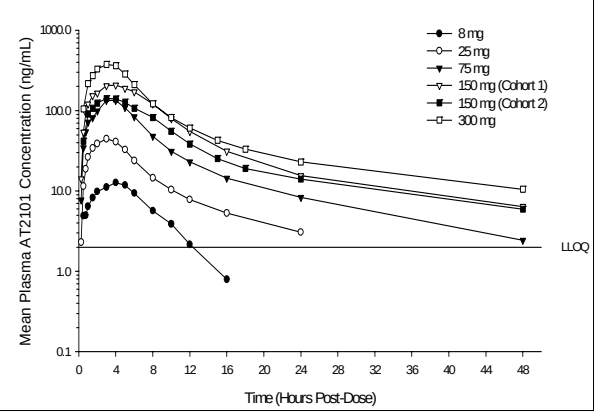
Mean Plasma AT2101 Concentration (ng/mL)
Time (Hours Post-Dose)
8 mg
25 mg
75 mg
150 mg (Cohort 1)
150 mg (Cohort 2)
300 mg
LLOQ

Figure 2. Dose Proportionality: AUC and C_{max} vs. Dose following a single oral dose

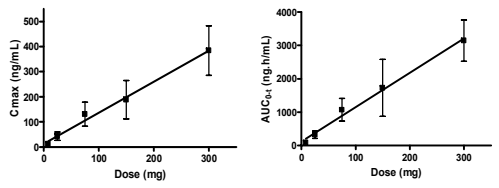
Cmax (ng/mL)
AUC0-t (ng.h/mL)
Dose (mg)

Summary

- **Median t_{max} values ranged from 3.00 to 3.50 hours**
- **Mean elimination half-life ranged from 5.14 to 19.9 hours; the greatest confidence in the results is at doses 75 mg and above where the half-life ranged from 16.1 to 19.9 hours.**
- **Mean CL/F values ranged from 31.8 to 48.6 L/h. There was no trend in CL/F values as dose increased, suggesting dose proportional increases in PK parameters**
- **AUC_{0-t} and C_{max} values for AT2101 appeared to be dose proportional (Fig. 2) (95% confidence limit of dose proportionality slopes included the value 1). This was true using both the combined 150 mg cohort results and each of the 150 mg cohorts separately in the linearity analysis.**

Pharmacokinetics: Multiple Ascending Dose

Table 5. Pharmacokinetic Results for AT2101 in Plasma and Urine on days 1 and 7 of a 7-day qd oral administration

Day 1

PK Parameters	Dose (mg)		
	25	75	225
AUC_{inf} (ng·h/mL)	278 (34)	1282 (48)	2235 (24)
C_{max} (ng/mL)	28.8 (49.1)	152.9 (66.0)	270.4 (28.2)
t_{max} (h)	3.50 (2.00 - 5.00)	4.50 (2.00 - 8.00)	3.00 (2.00 - 4.00)
$t_{1/2}$ (h)	10.6 (4.6)	7.9 (1.1)	11.1 (5.0)
CL/F (L/h)	46.6 (16.7)	31.7 (14.9)	51.0 (11.8)
V_{area}/F (L)	703 (441)	365 (189)	804 (348)
F_e (%)	8.8 (4.8)	20.2 (8.5)	13.1 (3.8)

AUC and C_{max} results are geometric mean (CV%), t_{max} results are median (min-max). All other results are arithmetic mean ± SD

Day 7

PK Parameters	Dose (mg)		
	25	75	225
AUC_{τ} (ng·h/mL)	296 (16)	1367 (34)	2620 (9)
C_{max} (ng/mL)	32.7 (32.3)	160.0 (48.4)	318.7 (11.5)
t_{max} (h)	2.00 (1.00 - 4.00)	3.00 (2.00 - 5.00)	3.50 (2.00 - 5.00)
$t_{1/2}$ (h)	13.2 (7.8)	17.9 (5.9)	16.5 (3.9)
C_{min} (ng/mL)	4.2 (1.1)	16.0 (4.8)	31.1 (4.3)
CL/F (L/h)	42.2 (6.6)	28.4 (8.9)	42.7 (3.6)
V_{area}/F (L)	767 (339)	761 (409)	1016 (247)
F_e (%)	12.1 (6.6)	21.6 (8.3)	16.3 (3.0)
Accumulation Ratio	1.39 (0.61)	1.20 (0.32)	1.34 (0.24)

Figure 3. Mean plasma concentrations of AT2101 during 7-day qd oral administration

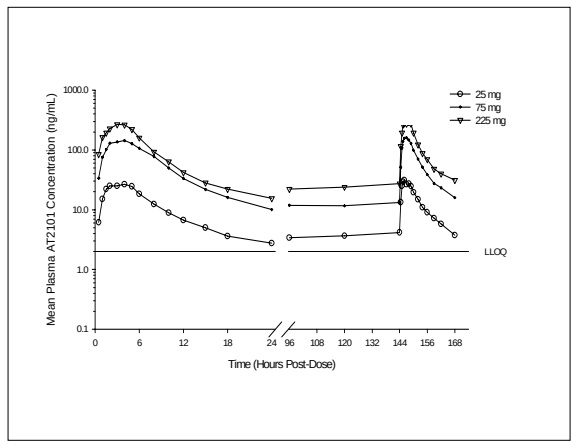
Mean Plasma AT2101 Concentration (ng/mL)
Time (Hours Post-Dose)
25 mg
75 mg
225 mg
LLOQ

Figure 4. Dose Proportionality: AUC and C_{max} vs. Dose following repeated daily oral doses

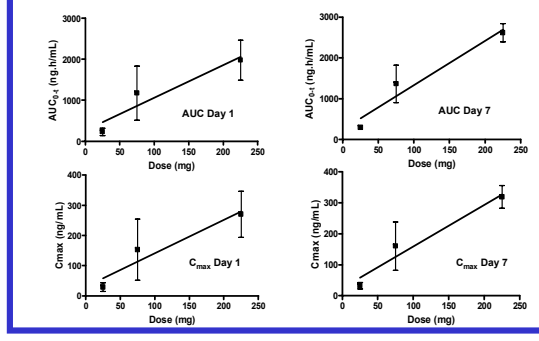
AUC Day 1
AUC Day 7
Cmax Day 1
Cmax Day 7
AUC0-t (ng.h/mL)
Cmax (ng/mL)
Dose (mg)

Summary

- **Following oral administration, the plasma concentrations of AT2101 declined in a biphasic manner.**
- **On day 1, median t_{max} values ranged from 3.0 to 4.5 hours; mean elimination half-life ranged from 7.9 to 11.1 hours. On day 7, median t_{max} values ranged from 2.0 to 3.5 hours.**
- **No trends were observed in CL/F or CL_R values; mean volume of distribution ranged from 365 to 804 L on day 1 and 761 to 1016 L on day 7.**
- **Accumulation ratios of 1.39, 1.20, and 1.34 were determined at the 25, 75, and 225 mg dose levels respectively. This accumulation is consistent with the observed plasma elimination half-life.**
- **AUC and C_{max} for AT2101 on both days 1 and 7 appeared to be dose proportional (Fig. 4) (95% confidence limit of dose proportionality slopes included the value 1).**
- **After multiple dose administrations from day 1 through 7, the PK of AT2101 was linear over the dose range of 25 to 225 mg, and steady state was attained within 5-days.**

Pharmacodynamics: GCase Activity in White Blood Cell Lysates

Figure 5. GCase activity in white blood cell lysates during repeated daily oral doses of AT2101 for 7 days followed by a 14-day wash-out period.

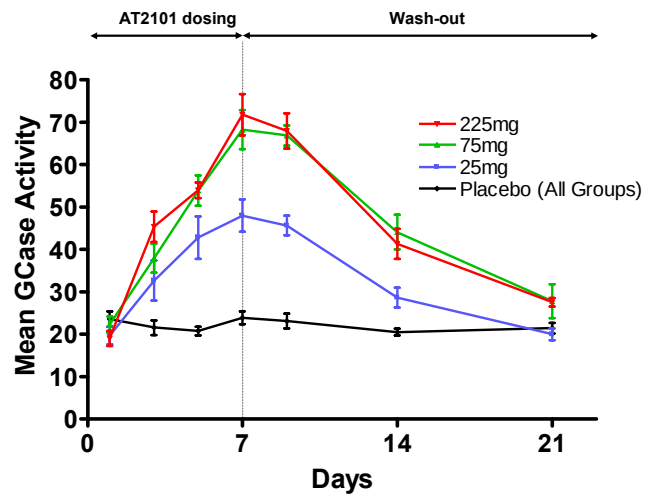
AT2101 dosing
Wash-out
Mean GCase Activity
Days
225mg
75mg
25mg
Placebo (All Groups)

GCase activity is reported as nmole 4MU / mg protein / hour
Activity values are mean ± SEM

PD Summary

- **GCase activity was measured in lysates of white blood cells isolated from subjects receiving AT2101 in daily oral doses for 7-days. Measurements were made pre-dose on days 1, 3, 5 and 7. Subjects returned for follow-up measurements on days 9, 14 and 21 during wash-out of AT2101.**
- **GCase activity was significantly increased in all treatment groups when compared to placebo.**
- **GCase activity increased with time in all treatment groups during administration of AT2101.**
- **Upon discontinuation of AT2101, GCase activity declined, returning to or near to baseline by day 21 (14 days of wash-out).**
- **The half-life for decline of GCase activity upon removal of AT2101 appeared to be about 4 to 5 days.**
- **GCase activity increased with increasing dose. The increase was most marked, in absolute terms, between 25 and 75 mg.**
- **Relative percent increases at day 7 (time of maximal increase) compared to baseline were 147%, 209% and 279% at 25, 75 and 225 mg respectively.**

Summary and Conclusion

- **The safety, pharmacokinetics, and pharmacodynamics of AT2101 were evaluated in two phase 1 studies in healthy volunteers.**
- **AT2101 was well tolerated at all doses.**
- **The combined results from both studies give an average half-life of 14 hours and t_{max} of 3.5 hours.**
- **AT2101 has linear pharmacokinetics (dose and time independent) and steady state was attained by day 5.**
- **Interpolation of C_{max} linearity data suggest a dose of 100 mg would provide a C_{max} of approximately 1 µM.**
- **AT2101 administration to healthy volunteers results in a dose-related and time-dependent increase in GCase activity in white blood cell lysates. An increase of near to 3-fold was observed at a daily dose of 225 mg for 7 days.**
- **Elevated GCase activity declines upon discontinuation of AT2101. The half-life for decline is about 4 to 5 days.**
- **AT2101 appears to act as a pharmacological chaperone for GCase. We hypothesize that even in healthy volunteers, AT2101 is able to bind and stabilize protein that would otherwise be degraded by the cellular quality control apparatus.**
- **These results support the further investigation of AT2101 as a pharmacological chaperone for the treatment of patients with Gaucher disease.**

American College of Medical Genetics
Nashville, Tennessee
March 2007